DEVELOPMENT OF A FROZEN FORMULATION OF ALLOCETRA-OTS, AN INNOVATIVE MACROPHAGE REPROGRAMMING CELL-BASED THERAPY

Veronique Amor-Baroukh[1], Moran Fattal[1], Omri Bezalel[1], AlinaSmush[1], Pnina Dekel[1], Yarden Dagan[1], Sivan Kagan[1], Chen Ankri[1], Iris Tavor[1], Dror Mevorach[2], Oren Hershkovitz[1]

[1]Enlivex Therapeutics, Ness Ziona, Israel; [2] Hadassah-Hebrew University, Jerusalem, Israel

ENLIVEX
immune rebalancing

Introduction

Enlivex Therapeutics Ltd. is developing **Allocetra-OTS, an allogeneic cell therapy,** consisting of mononuclear cells induced to an **early, stable apoptotic state.** Allocetra-OTS harnesses the naturally occurring activity of apoptotic cells to induce an immuno-modulated state through **reprogramming of non-homeostatic macrophages**.

The 1st generation drug product was a fresh, liquid formulation (LDP) of Allocetra-OTS cells in Ringer's lactate ,with a shelf-life of 96h at 2-8°C. The 2nd generation frozen formulation was developed (FDP) and necessary adjustments were introduced to the manufacturing process.

The final product is a cell suspension of Allocetra-OTS cells in Plasma-Lyte and CryoStor5®, provided as a ready-to-use therapy which can be stored at clinical sites with immediate applicability. The FDP long-term stability allows full sterility release prior to administration to patients, and therefore improves the overall control of product quality.

The comparability assessment strategy relied on the fact that the manufacturing processes of LDP and FDP are nearly identical up to the final formulation step.

Objective

Development and implementation of a frozen formulation of Allocetra-OTS, to support expansion of clinical studies into EU/US and additional countries, commercialization, and improve the overall product control strategy.

Conclusion

- ❖ The **frozen formulation of Allocetra-OTS** was developed to **improve and facilitate its clinical utility and commercialization**.
- ❖ The thorough **comparability** exercise demonstrated **equivalence in most quality attributes** and **all release tests met the acceptance criteria**.
- ❖ The accumulated data from process development studies, analytical comparability study, *in vivo* pharmacology and toxicology studies, and the potential for immediate applicability for the patient, all supported the implementation of frozen Allocetra-OTS in clinical programs.
- ❖ The frozen formulation was **approved by regulatory agencies** in ongoing clinical studies in **sepsis** (NCT04612413) and **cancer** (NCT05431907 and NCT05581719), and in early phase clinical studies in new indications.

Methods

The strategy for the development and implementation of the new formulation relied on regulatory guidelines and industry practices. The comparability was assessed based on **ICH Q5E guideline and EMA Q&A document on Comparability Considerations for ATMPs (EMA/CAT/499821/2019),** implementing a stepwise, risk-based approach as described below:

- ❖ **Process Development Experiments** –A thorough risk assessment was performed, based on accumulated data and knowledge, to assess the main risks of the formulation and process changes, and their potential impact on the safety and efficacy of Allocetra-OTS final product.
- ❖ **Analytical Comparability exercise** – The comparability study was designed based on accumulated data from the abovementioned activities and compared the product quality attributes including purity, impurity, potency, process performance ,stability, and additional characterization studies.
- ❖ ***In vivo* pharmacology / GLP Toxicology studies** – Several *in vivo* studies were performed using both LDP and FDP. Moreover, a GLP toxicology study was performed using FDP batches.
- ❖ **Stability Studies** – Preliminary stability studies were performed using process development batches. In addition, comparability batches were placed under a long-term stability program. Post-thaw stability studies were performed as part of process development.

Results

Cryopreservation formulation optimization. As part of the development of the frozen formulation of Allocetra, several parameters were evaluated: resuspension medium, cryopreservation agent, DMSO content, various excipients, cell concentration, product volume, and more (data not shown).

Figures A and B demonstrate the effect of DMSO source and freezing formulation on apoptosis and late apoptosis levels, respectively, following thawing of the final product.

Intra-batch comparison of Allocetra liquid and frozen formulations.

For the comparability exercise, batches of FDP and LDP were manufactured from the same cell source (Figures C – E).

FDP and LDP samples were simultaneously tested at two timepoints (LDP T=17h and T=90h).

Both FDP and LDP formulations demonstrated equivalence in statistical testing for the following parameters: purity / impurity (CD45+ / CD45- cells), apoptosis level, cell populations composition (not shown), and potency.

Equivalence was not shown for late apoptosis level, yet results were within specifications.

In addition, the FDP was thoroughly assessed by *in vivo* pharmacology and a GLP repeated dosing toxicology study, both of which support a similar safety and efficacy profile of the two formulations (data not shown).

A

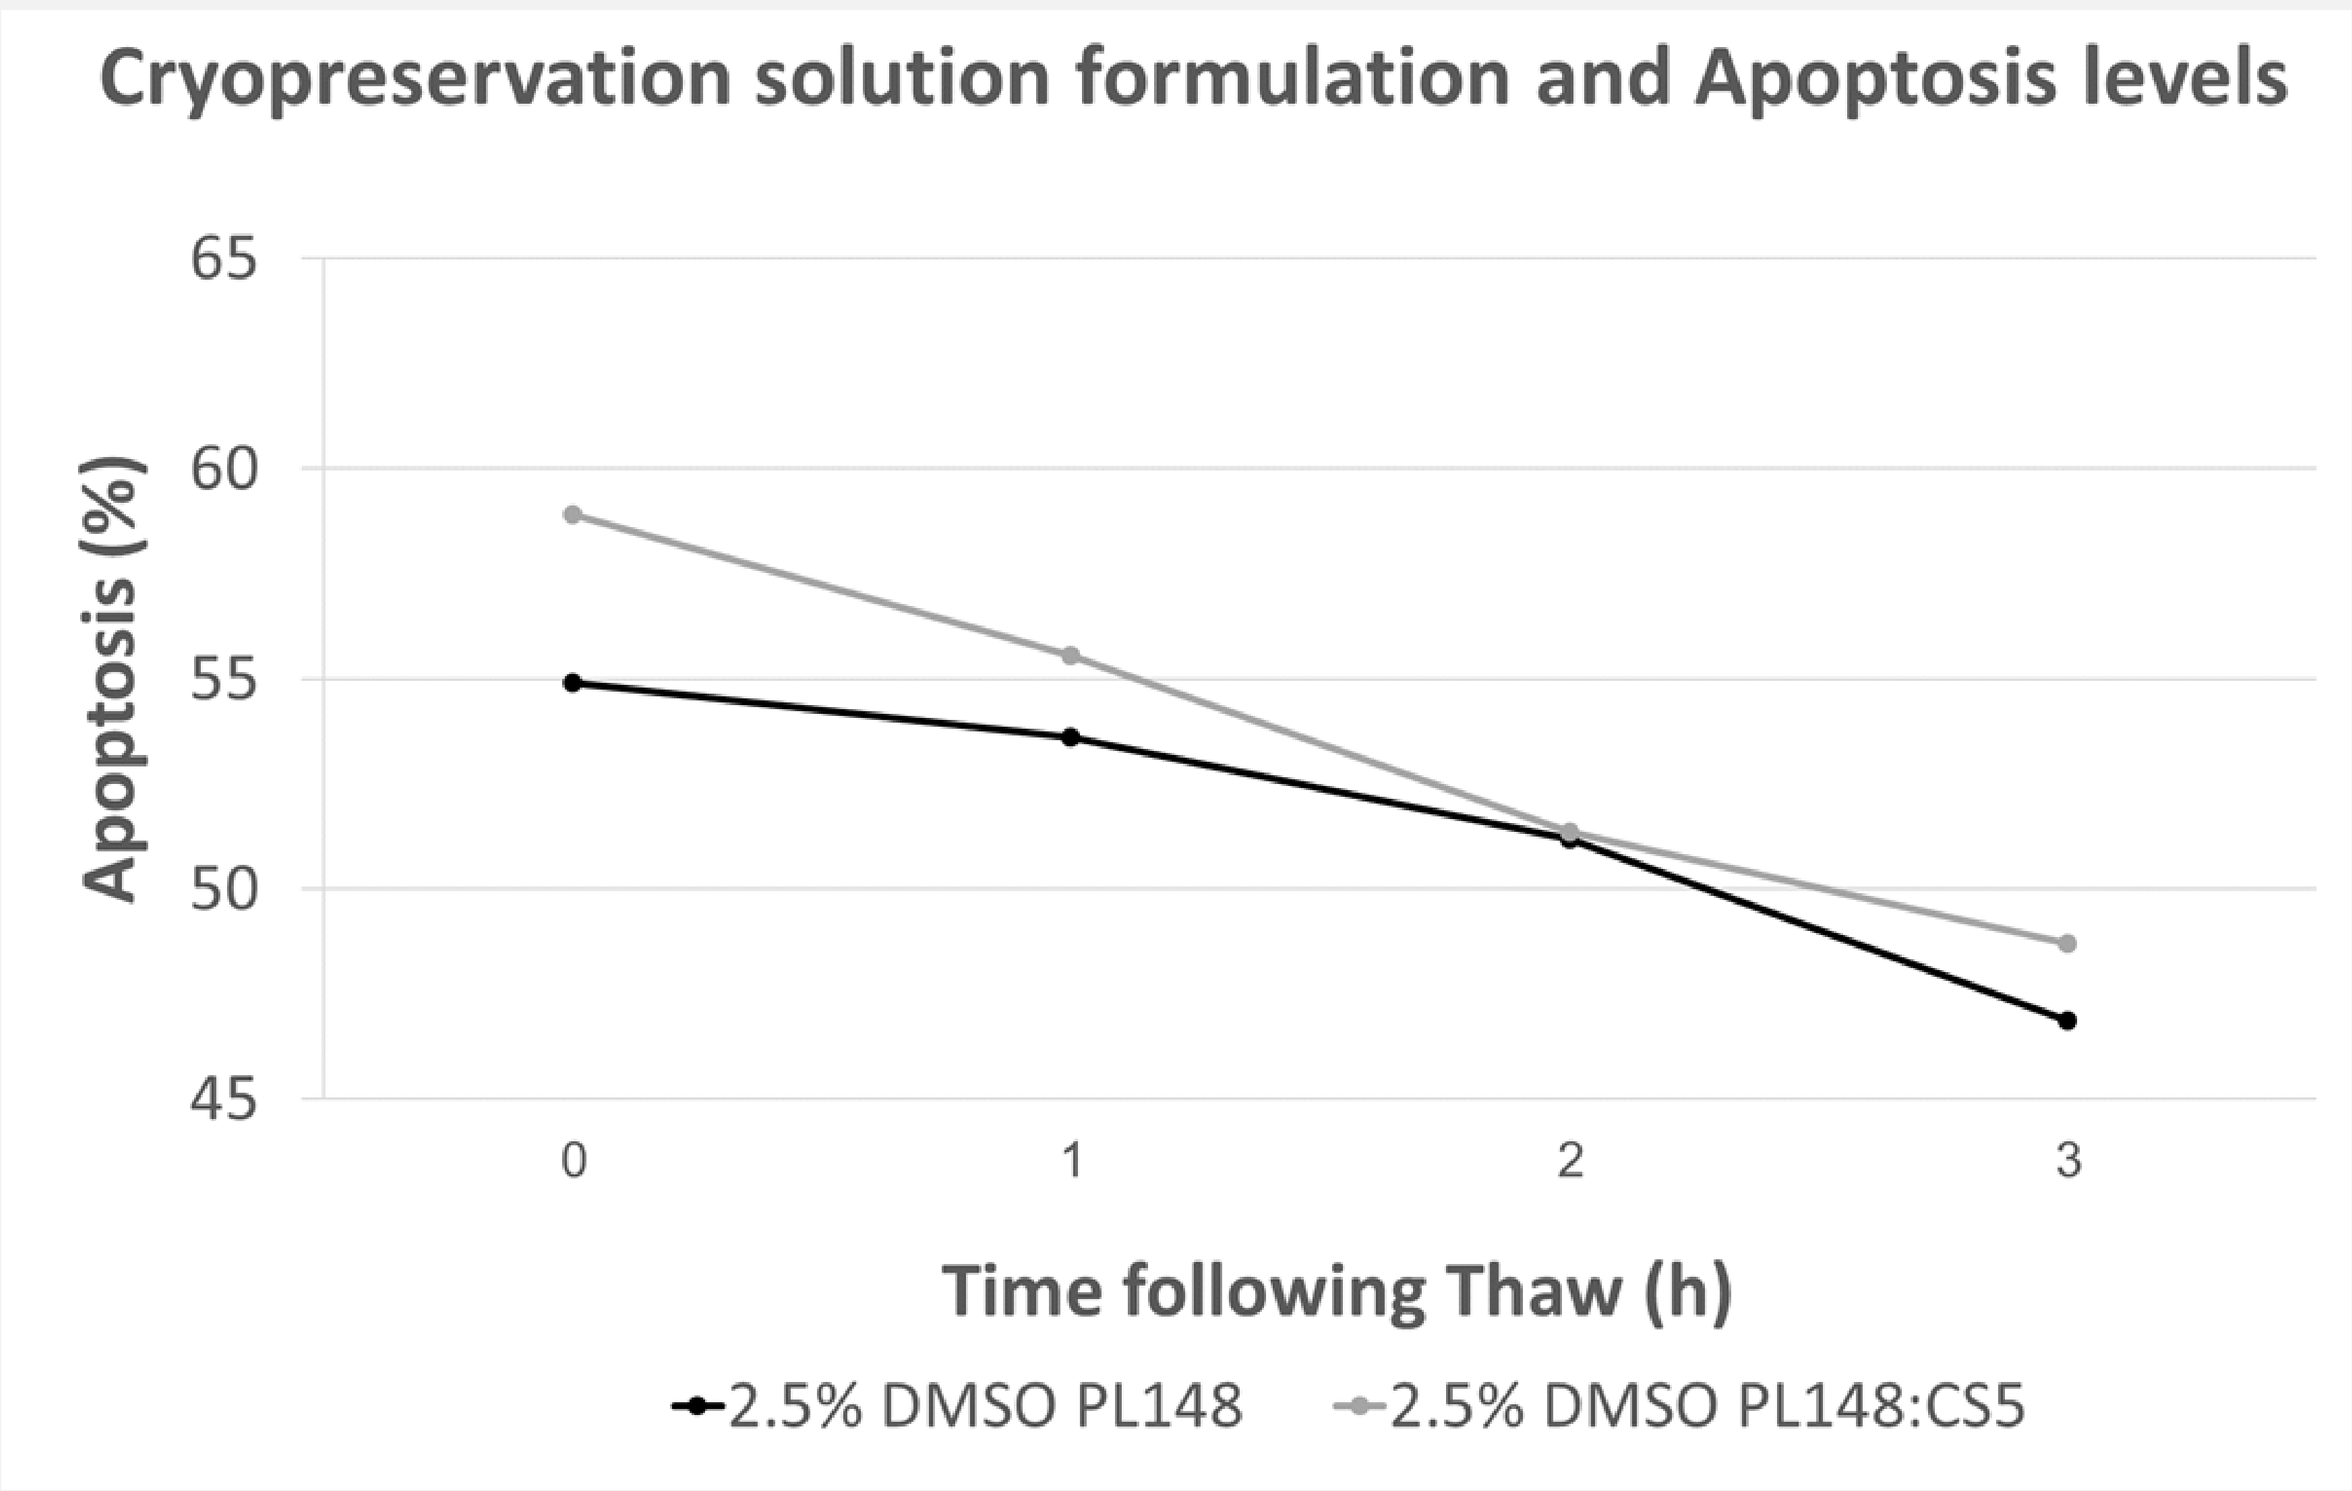
Cryopreservation solution formulation and Apoptosis levels
Apoptosis (%)
65
60
55
50
45
0
1
2
3
Time following Thaw (h)
2.5% DMSO PL148
2.5% DMSO PL148:CS5

B

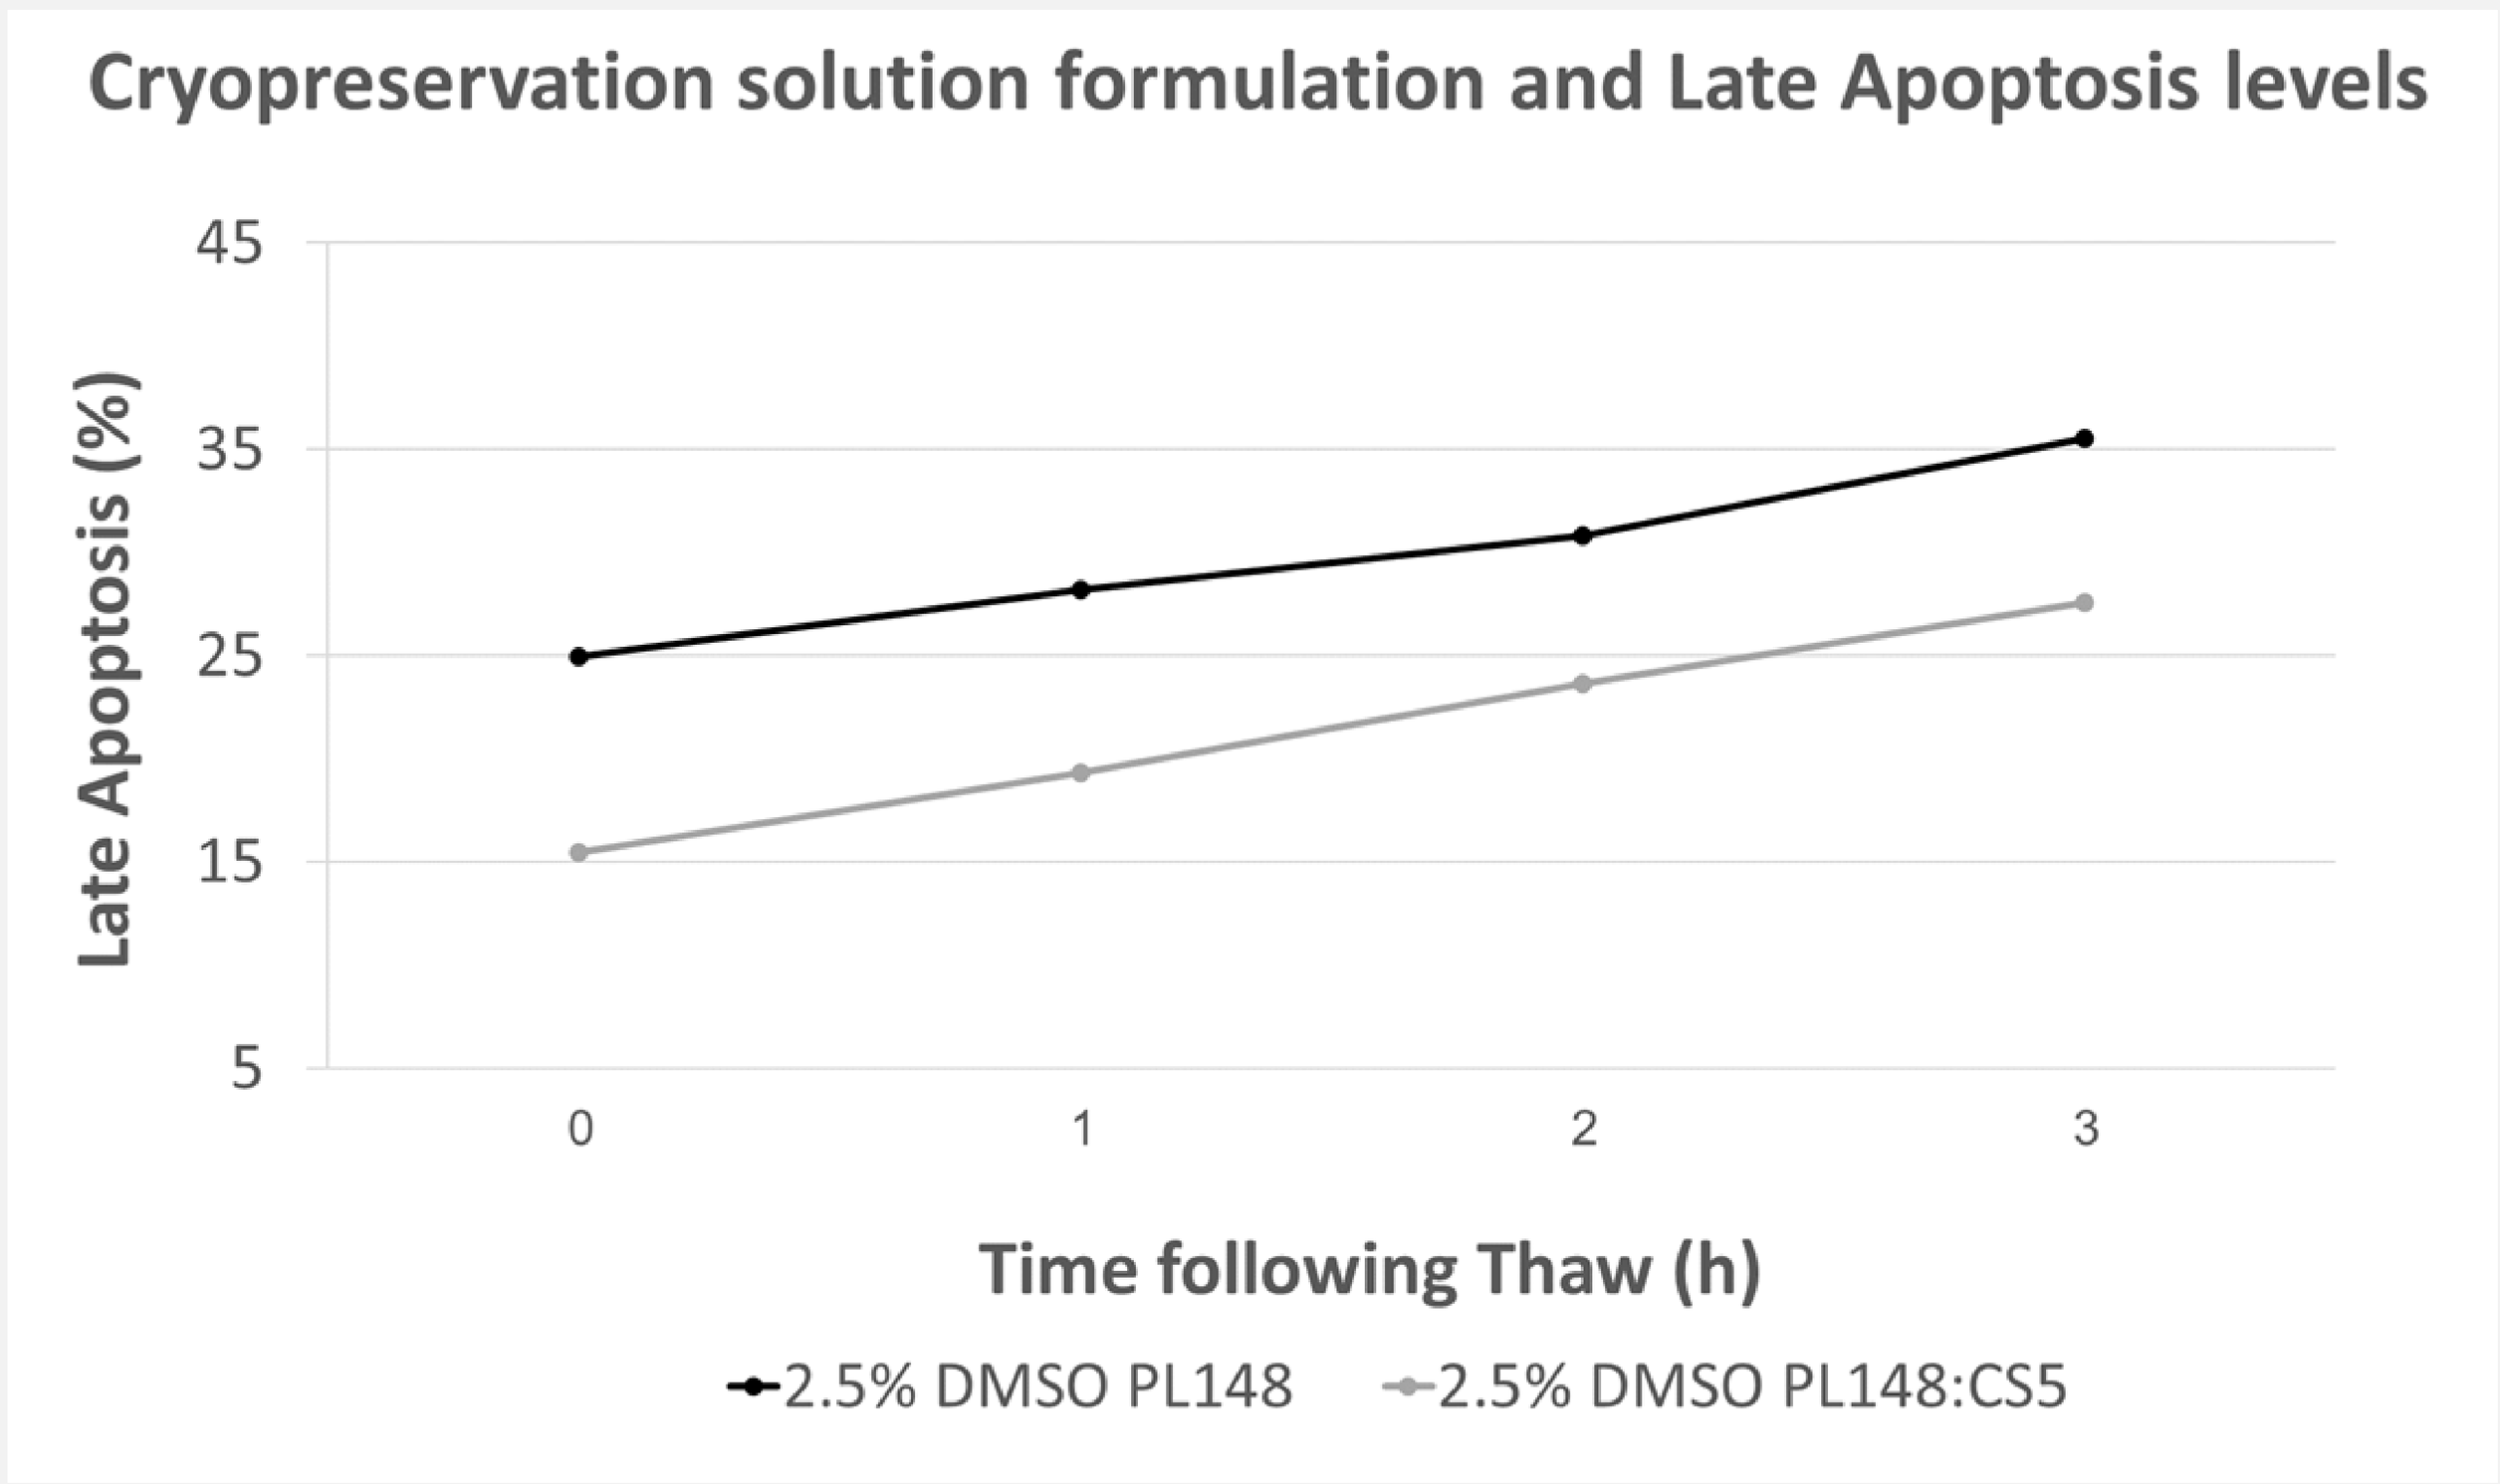
Cryopreservation solution formulation and Late Apoptosis levels
Late Apoptosis (%)
45
35
25
15
5
0
1
2
3
Time following Thaw (h)
2.5% DMSO PL148
2.5% DMSO PL148:CS5

C

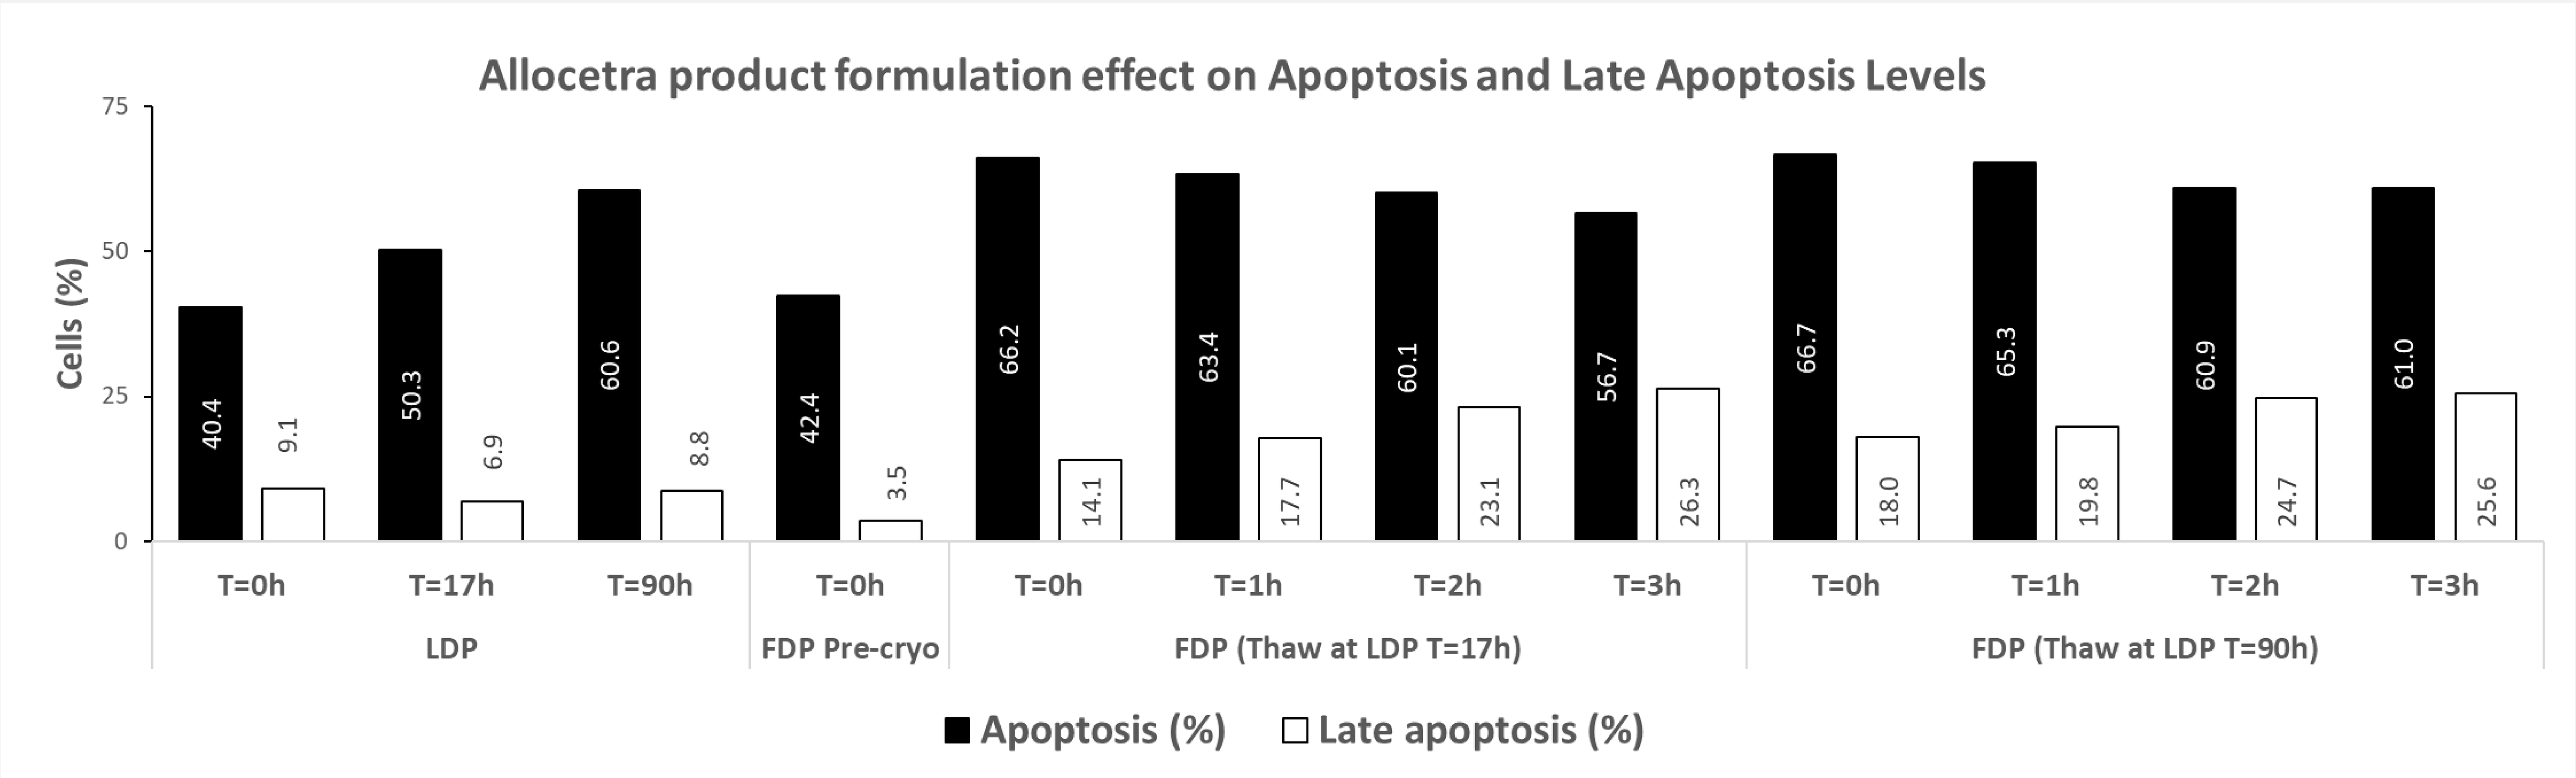
Allocetra product formulation effect on Apoptosis and Late Apoptosis Levels
Cells (%)
75
50
25
0
40.4
9.1
50.3
6.9
60.6
8.8
42.4
3.5
66.2
14.1
63.4
17.7
60.1
23.1
56.7
26.3
66.7
18.0
65.3
19.8
60.9
24.7
61.0
25.6
T=0h
T=17h
T=90h
LDP
T=0h
FDP Pre-cryo
T=0h
T=1h
T=2h
T=3h
FDP (Thaw at LDP T=17h)
T=0h
T=1h
T=2h
T=3h
FDP (Thaw at LDP T=90h)
Apoptosis (%)
Late apoptosis (%)

D

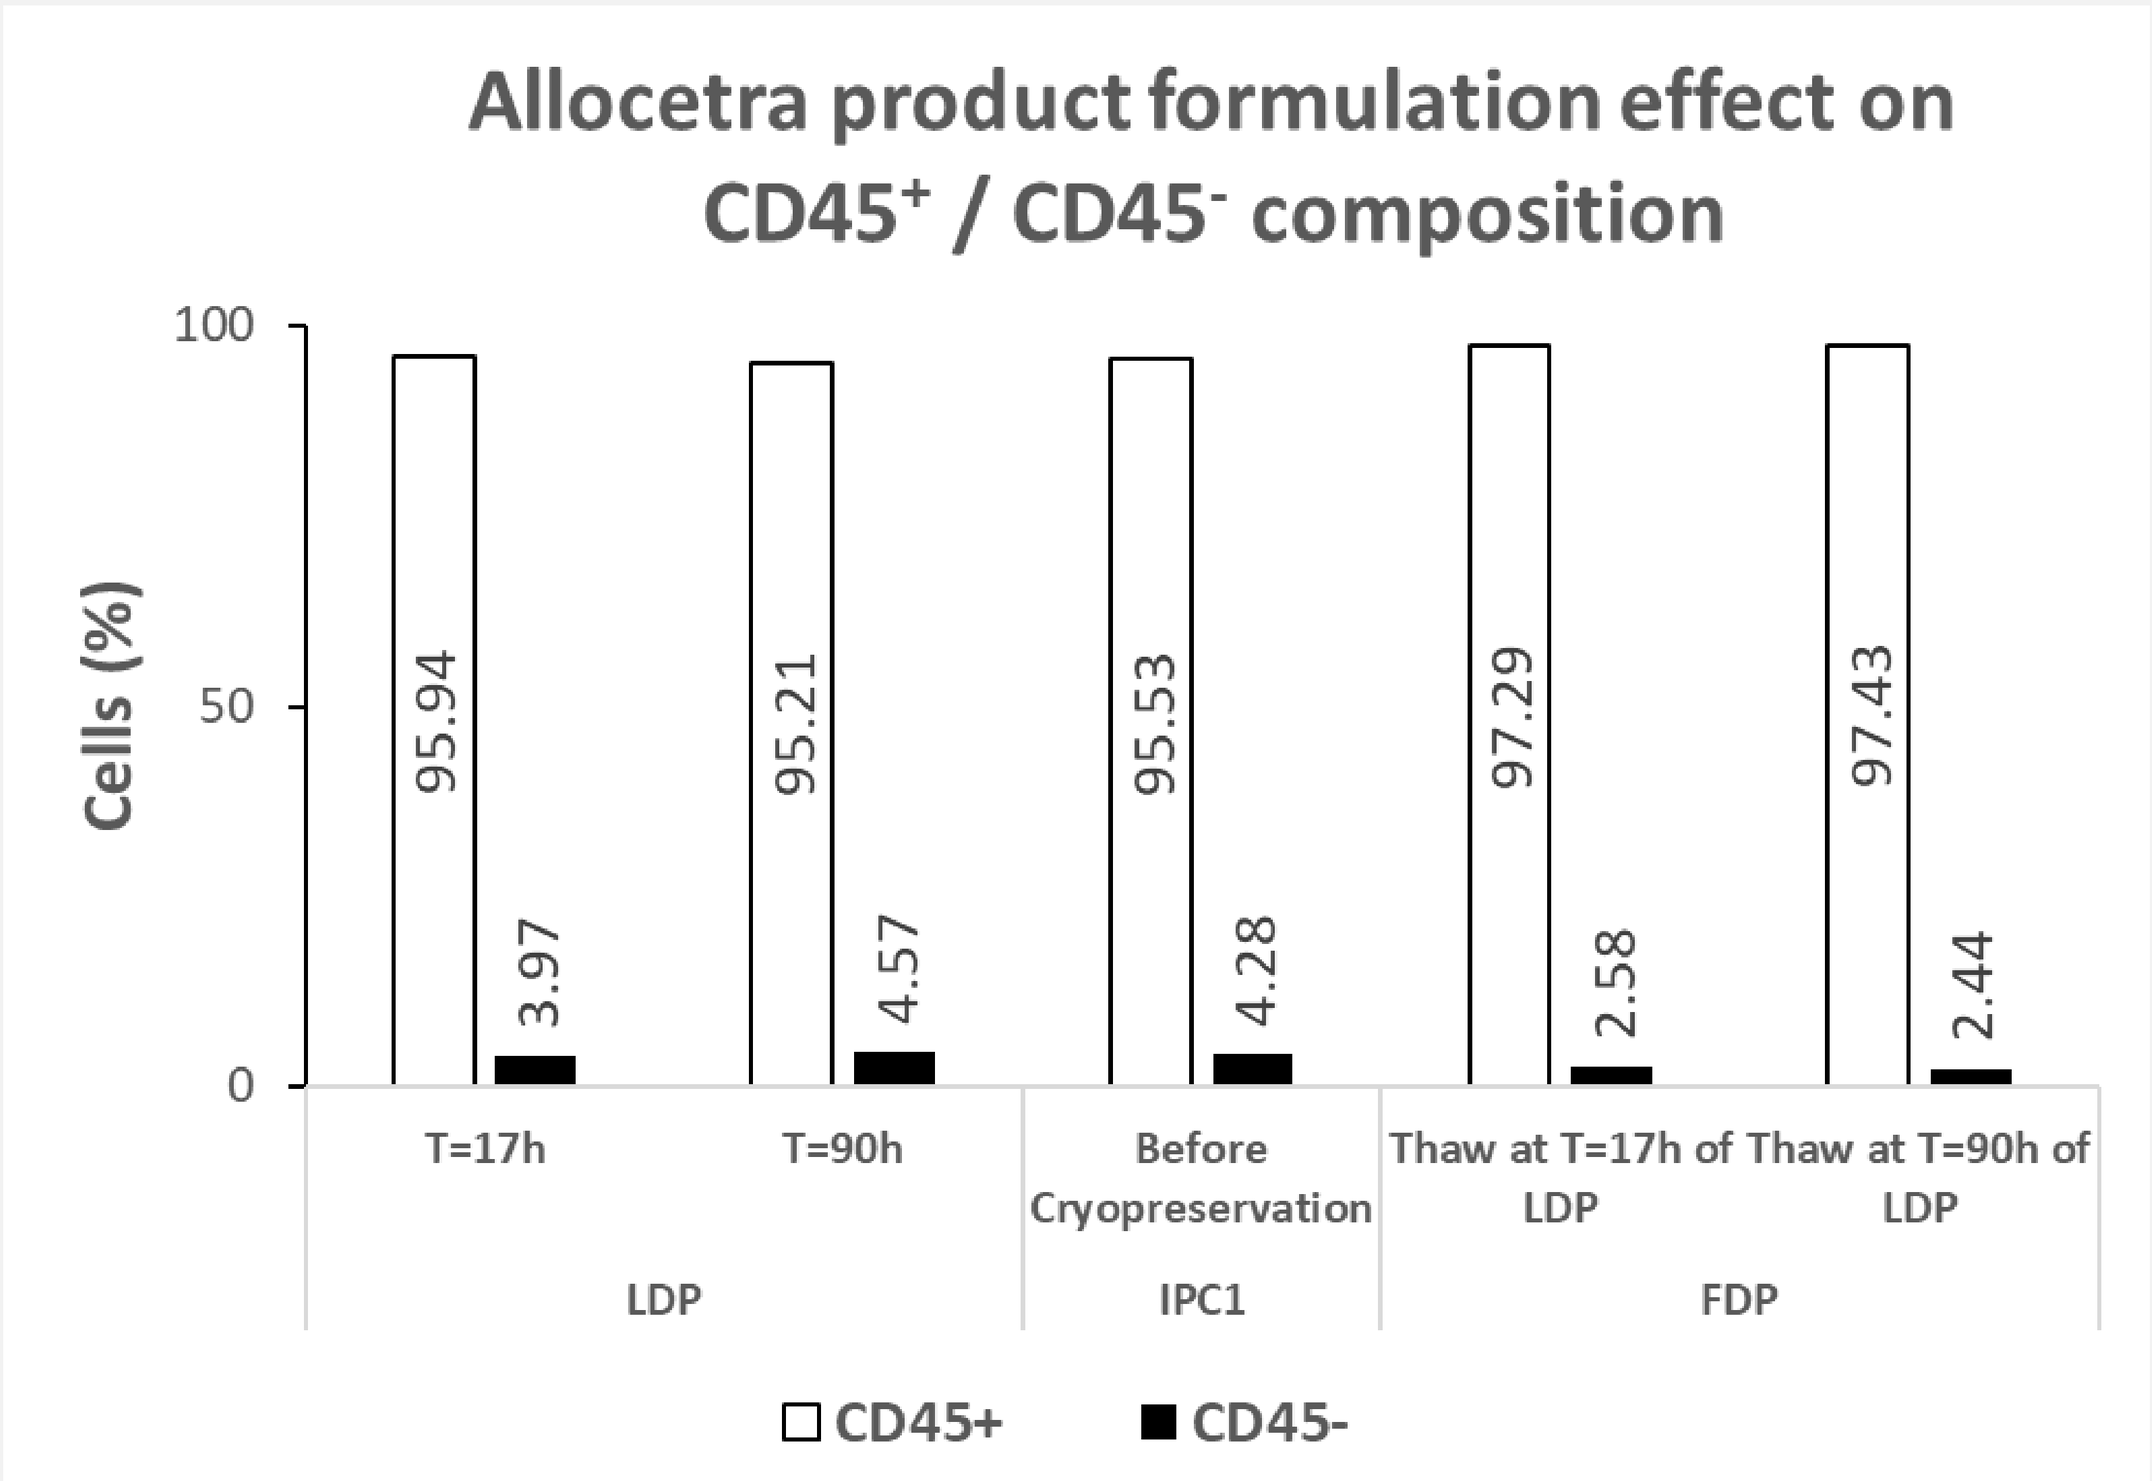
Allocetra product formulation effect on CD45+ / CD45- composition
Cells (%)
100
50
0
95.94
3.97
95.21
4.57
95.53
4.28
97.29
2.58
97.43
2.44
T=17h
T=90h
LDP
Before Cryopreservation
IPC1
Thaw at T=17h of LDP
Thaw at T=90h of LDP
FDP
CD45+
CD45-

E

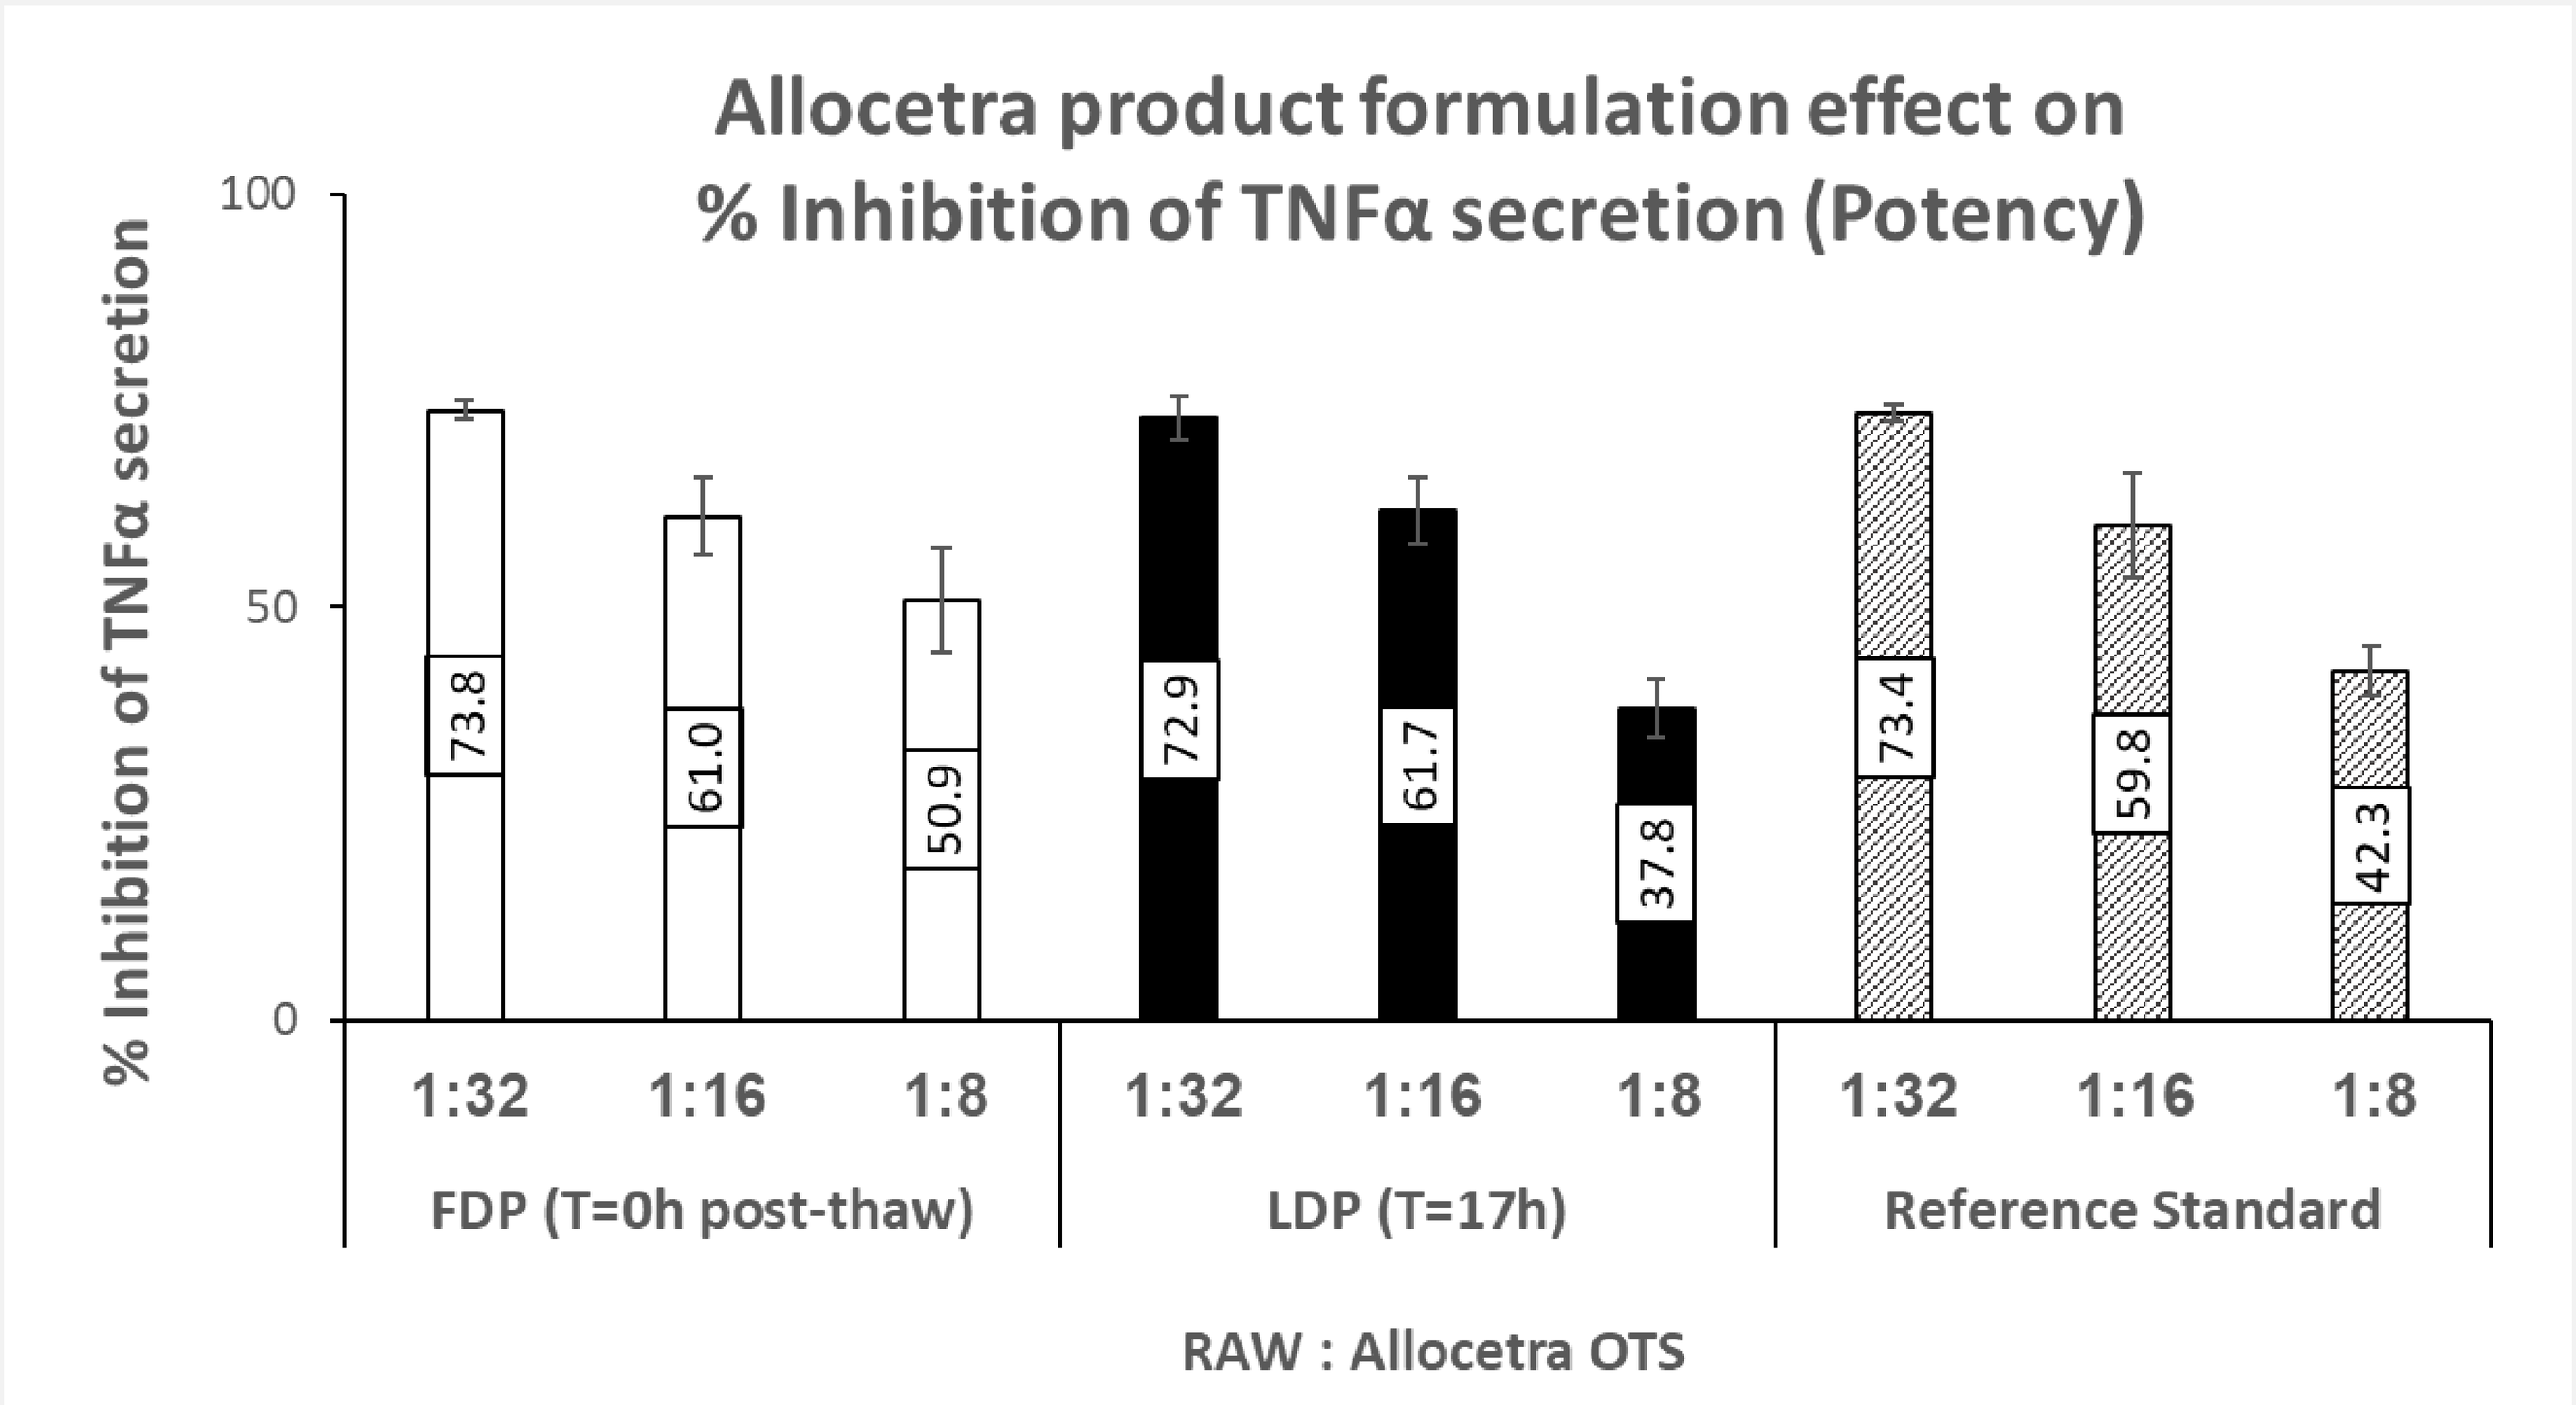
Allocetra product formulation effect on % Inhibition of TNFα secretion (Potency)
% Inhibition of TNFα secretion
100
50
0
73.8
61.0
50.9
72.9
61.7
37.8
73.4
59.8
42.3
1:32
1:16
1:8
FDP (T=0h post-thaw)
1:32
1:16
1:8
LDP (T=17h)
1:32
1:16
1:8
Reference Standard
RAW : Allocetra OTS